Mirroring Disease Progression and Executive Functioning in MS-patients by DTI-based Fiber Tracking and Various Brain Atrophy Measures

F. Fink[1,6], M. Butt[1], J. Klein[4], M. Lanz[1], M. Lentschig[3], T. Mitrovics[2], H. Hildebrandt[1,5]

(1) Dpt. of Neurology (CA Prof. Schwendemann), (2) Dpt. of Neuroradiology, Klinikum Bremen-Ost and (3) Klinikum Bremen-Mitte
(4) MeVis Research, Bremen, (5) Institute of Psychology, University of Oldenburg
(6) NICI, University of Nijmegen, the Netherlands

Background

During the last years various MRI scanning protocols assessing disease progress in MS have been developed. Measures of brain atrophy for example received considerable attention because they can easily be extracted from T1 MRI scans. But in MS patients, cognitive functions and especially executive functions do not always correlate with brain atrophy measures [1], which leads to the question whether the use of alternative MRI scanning protocols would improve this relation. Diffusion tensor imaging is a quite recent technique in MRI scanning that allows assessing the integrity of fiber structures in regions of interest and – by usages of appropriate mathematical models - also of whole fiber tracts. The integrity of fiber bundles is deduced from the degree of anisotropic diffusion along the tract. Because MS is an inflammatory and neurodegenerative disease, which leads to a demyelinisation of fibers and axonal injury, DTI promises to reflect the destructive consequences of disease progress. This is of distinguished relevance for studying cognitive impairments in MS and especially deficits in executive functions. Almost all higher cognitive functions rely on intracortical and subcortical networks and it is well-known that intact executive functions presuppose an intact fiber coupling of postcentral and frontal brain areas.

To explore the relevance of Diffusion Tensor Imaging in MS and to look for its potential contribution to the explanation of executive functions deficit in MS, we used DTI and measures of brain atrophy (brain parenchymal fraction, standar-dized volume of the second, third and fourth ventricel and of gray and white matter) to see whether DTI or brain atrophy explains better the

a) disease duration
b) clinical staging of the disease progress
c) performance in executive functioning

Methods

To investigate the concurrent validity of DTI and brain atrophy measure, we investigated 30 relapsing-remitting MS patients with MRI scanning with a neurological examination (Expanded Disability Status Scale, EDSS), clinical staging by the Multiple sclerosis functional composite score (MSFC) and a comprehensive neuropsychological test battery, which also included two experimental tests for executive functions, conditional responding and object alternation. The task, which the subject has to master during conditional responding, is to switch between two ways of responding in the face of a change of stimuli. During object alternation, the subject has to alternate the preferential object.

To analyze the relation between executive functions and fiber integrity, we focused on the superior longitudinal fascicle (SLF) which connects secondary and tertiary association centers in the parietal lobe with premotor and prefrontal areas of the frontal lobe. It has been argued that this fiber tract is of primary importance for updating representations as well as regulating and selecting actions [2].

	Mean	range
Age (years)	45,6	30-65
Disease duration (years)	8,6	0,5-28
EDSS	2,9	1,0-7,0
MSFC (z-score)	.0251	-1.44-.63

Table 1: Patients' characteristics.

Neuropsychological examination:

- Working memory: 2-back performance (TAP Arbeitsgedächtnis), PASAT (Paced auditory serial addition test)
- Executive functions: two experimental tasks concerning object alternation and conditional responding. Two additional tasks were included which control for unspecific impairments in the patients. Substraction scores were calculated in order to exclude global disease related impairments from EF dysfunctions.
- Verbal memory (CVLT)
- Sustained attention (TAP Alertness)
- Two subtests for verbal and for performance intelligence from the HAWIE-R
- Becks Depression Inventory, Fatigue Severity Scale, Quality of Life (SF12)

Neuroimaging

- Diffusion Tensor Imaging (see Figure 1 for two examples of fiber tracking).
- T1 weighted high resolution 3 D scans.
- T2 weighted 2 D scans.
- NeuroQLab for image processing and quantification of brain atrophy measures (from T1 scanning) and fiber tracking (from DTI scanning).

Quantification of fiber tracking was done by calculating the fractional anisotropy (FA) for a discrete number of position along the bundle. For that purpose, an average FA value was determined for each position, where a single FA can be computed from the eigenvalues (λ_1, λ_2, λ_3) of a diffusion ellipsoid:

$$FA = \sqrt{\frac{1}{2}} \frac{\sqrt{(\lambda_1 - \lambda_2)^2 + (\lambda_2 - \lambda_3)^2 + (\lambda_3 - \lambda_1)^2}}{\sqrt{\lambda_1^2 + \lambda_2^2 + \lambda_3^2}}$$

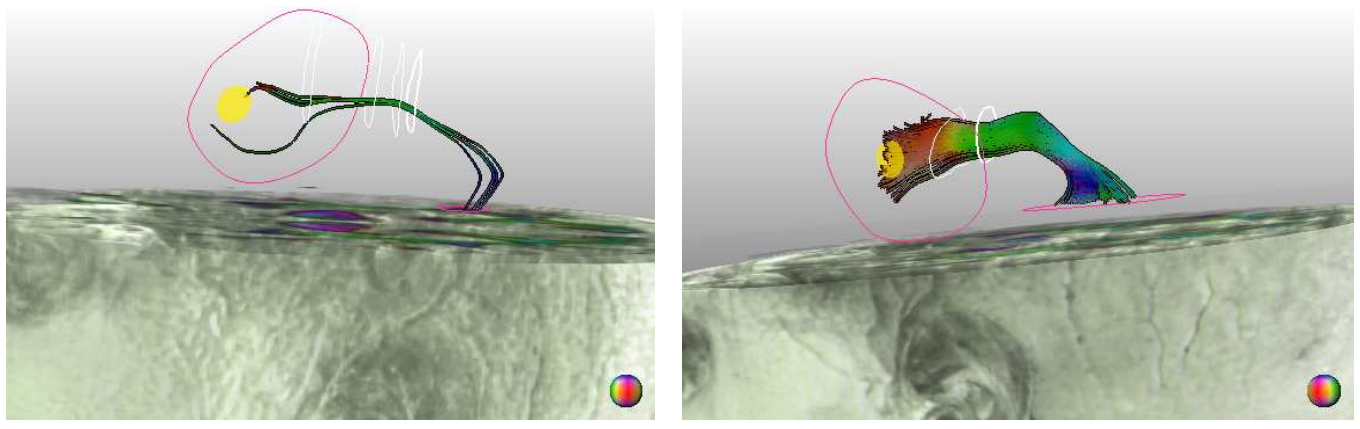

Figure 1: Two examples of the superior longitudinal fascicle (SLF). Determining the line of section on the same latitude as the splenium corporis callosi (from a coronal/axial viewpoint) 3, the green-coded area lateral to the pyramidal tract was defined as ROI and the fibers were tracked. Left: The diffusion process is disturbed by demyelination and axonal loss so that only few fibers can be reconstructed. Right: Example of a more intact fiber bundle.

Statistics

Regression analyses (method: forward regression) modeling the relationship between

- disease duration, clinical status and executive functioning on the one hand and
- SLF and brain atrophy (BPF, standardized 2nd, 3rd, 4th ventricles, white, grey matter) on the other
- using age as variable to control for disease independent parenchymal loss.

Results

- Both brain atrophy measures and the FA of the SLF correlated with symptom duration.
- Disease duration and EDSS score were predicted by lateral ventricle volume or BPF.
- SLF's anisotropy (especially of the left hemisphere) predicted the MSFC z-score. (R^2=.529; p=.005).
- The right SLF's anisotropy predicted performance on the object alternation task (R^2=-.396; p=.033).
- More specific results are expected when finishing the investigation of healthy controls.

Discussion

- Lateral ventricle volume and BPF are suitable markers for mirroring disease duration and neurological staging. They outperform fiber tracking of the superior longitudinal fascicle in this respect.
- Anisotropy of the SLF measures functional staging according to the MSFC and delivers and additional value for predicting executive function impairments (object alternation) above other brain atrophy measures.
- Therefore, fiber tracking seems to be the better method to analyze specific and especially cognitive function loss in MS, whereas brain atrophy measures offer an easily to be extracted predictor for global disease progression.

We conclude that Diffusion Tensor Imaging represents a new and exciting research tool in investigating inflammatory and neuro-degenerative diseases and we expect that it will have a considerable impact on studies investigating the structural architecture of the brain when performing executive functions.

This study was sponsored by Sanofi-Aventis, Bayer Health Care and Merck-Serono

References:

1. Lanz, M., Hahn, H.K., & Hildebrandt, H. (2007). Brain atrophy and cognitive impairment in multiple sclerosis: a review. *Journal of Neurology, 254(2),* 43-48
2. Petrides, M., & Pandya, D.N. (2002). Association pathways of the prefrontal cortex and functional observations. In D.T. Stuss & R.T. Knight (Eds.). *Principles of frontal lobe function* (pp. 31-50). New York: Oxford University Press.
3. Mori, S., Wakana, S., van Zijl, P.C.M., & Nagae-Poetschner, L.M. (2005). *MRI atlas of human white matter*. Elsevier.